Bayesian Modeling of QT Interval: Focus on Baseline Measurements

Ihab G. Girgis†, Jeremy Hing††, Partha Nandy††, Filip De Ridder†, and Surya Mohanty†

Johnson & Johnson Pharmaceutical Research & Development

† Advanced Modeling & Simulation, Biometrics and Clinical Informatics

†† Advanced Modeling and Simulation, Clinical Pharmacology

Introduction

Detecting drug-induced effects on cardiac repolarization, measured by the length of the QT interval on an ECG, is a closely monitored safety element in drug development. More recently, it is thoroughly scrutinized in regulatory submissions. Baseline QT intervals can be influenced by a number of factors, such as, heart rate (HR), administration of placebo, gender, and natural circadian rhythm. In addition, there might be other unknown factors making it highly variable across population and the analysis of such data is complex. Accurate modeling of baseline QT becomes an important first step in evaluating effects of drugs. Changes to this baseline model after the administration of the investigational drug will reflect the effect on the QT/QTc interval.

Objective

The objective of this work is to model the baseline QT data in healthy subjects using a hierarchical Bayesian approach and to explore the influence of gender, RR interval (RR= 60/HR), and circadian rhythm on the QT interval. Also, the effect of placebo was tested.

Data

Data was obtained from a double-blind, randomized, placebo- and positive-controlled, 3-way crossover study in 40 healthy subjects (22 M, 18 F, age 30 ± 8 years, BMI 24 ± 2). Only drug-free data (baseline and placebo data) was used for this analysis. For each subject, 18 pre-specified time points were obtained from13 hour continuous 12-lead ECG recordings. The dataset used in this analysis contains 2160 time points for baseline with 6 QT missing values and 720 time points for placebo. The clock time data was imputed from the time relative to dosing and dosing window.

Data Exploration

Data pooled by treatment groups and gender showed a slight shortening of QT interval, by about 3 ms (at RR=1 sec), during placebo treatment as compared to baseline (Figure 1). Women consistently exhibit a longer QT interval than men by about 15 ms (at RR=1 sec). As heart rate increases the mean offset between men and women seems to decrease (Figure 2). The Fredericia-corrected QT interval (QTcF) for all pooled data shows a circadian variation of about 5-10 ms, peaking during morning hours (Figure 3). The same pattern is observed at the individual level (Figure 4).

Figure 1. QT Interval versus RR Interval for Different Treatment Groups

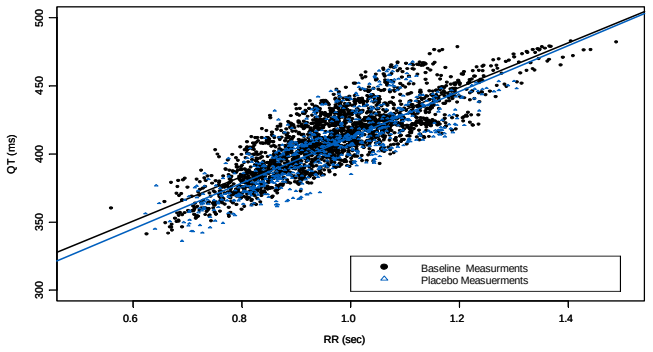
QT (ms)
RR (sec)
Baseline Measurments
Placebo Measuerments

Figure 2. Baseline QT Interval versus RR Interval by Gender

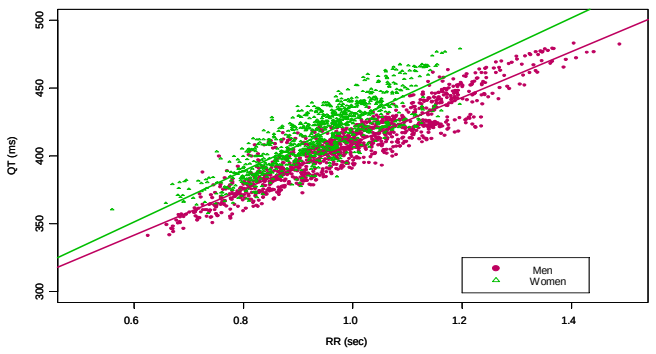
QT (ms)
RR (sec)
Men
Women

Figure 3. Circadian Rhythm Trend Line for Baseline Pooled Data

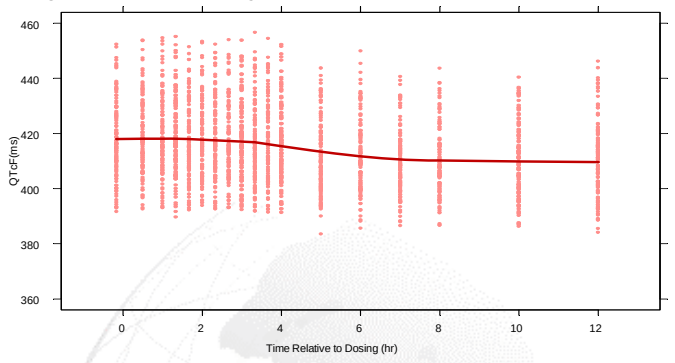
QTcF(ms)
Time Relative to Dosing (hr)

Figure 4. Circadian Rhythm Trend Line for Baseline Individual Data

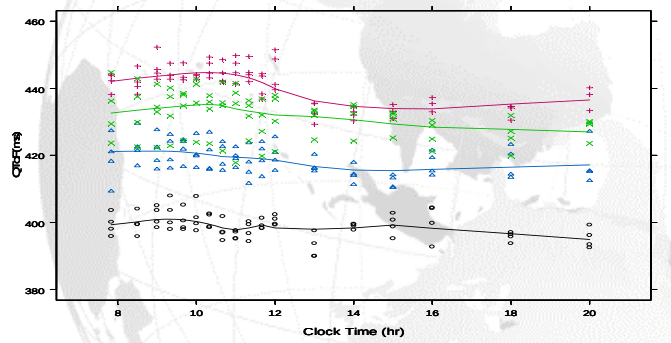
QTcF(ms)
Clock Time (hr)

Methods

A fully Bayesian approach using Markov chain Monte Carlo method was employed for mixed-effects model building using WinBUGS©[1]. GridBUGS interface (Girgis et al.[2]) was used for parallel computation and multi-chain runs. The hierarchical structural model for QT interval is presented as follows:

$$QT_{ij} = \left(QTc_i + Cir_i + \theta_{Pi} * Pla_i + \theta_{Gi} * Gen_i\right) \bullet Corr_i \bullet (1+\varepsilon_{ij}) \quad \text{Equation 1}$$

The model is comprised of four sub-models: placebo effect, gender effect, heart rate, and circadian rhythm[3].

- QT_{ij}: j^{th} measured observation of the QT interval (ms) for the i^{th} individual
- QTc_i: typical value of the baseline QT for a male subject,
- Cir_i: circadian rhythm function (see below)
- θ_{pi}: typical value of the placebo effect
- Pla_i: indicator for the placebo treatment
- θ_{Gi}: typical value of the gender effect
- Gen_i: indicator for gender.
- ε_{ij}: residual normal error.

In the hierarchical structure model, the conditional probability distribution of QT_{ij} is assumed to be normal, Equation 2.

$$p(QT_{ij} \mid \theta_i, \sigma^2) \propto N(f(\theta_i; t_{ij}; P_i; S_i), \sigma^2) \quad \text{Equation 2}$$

- $f(.)$: Function of the individual parameters θ_i
- t_{ij}: Clock time (24 hours)
- P_i: Individual's placebo dosing record
- S_i: individual's gender
- σ^2: residual error variance

Individual parameters, θ_i, were assumed to have a multivariate normal distribution with a variance-covariance matrix, Ω, of between subject random variability. Ω^{-1} is modeled by Wishart distribution. Non-informative priors were used. Graphical representation of the hierarchical structure model is shown in Figure 5. Model comparison and selection were based on posterior predictive distributions, parameters autocorrelation, Brooks-Gelman-Rubin (BGR) diagnostic, and deviance information criterion (DIC)[3]. Two Markov chains were run with 35,000 samples each and 5,000 samples burn in.

Figure 5. Graphical Representation of the Hierarchical Structure Model

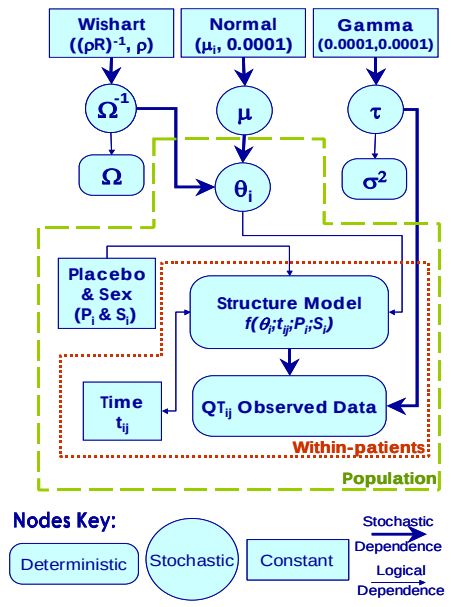
Wishart
((ρR)⁻¹, ρ)
Normal
(μi, 0.0001)
Gamma
(0.0001,0.0001)
Ω-1
μ
τ
Ω
θi
σ2
Placebo & Sex (Pi & Si)
Structure Model f(θi;tij;Pi;Si)
Time tij
QTij Observed Data
Within-patients
Population
Nodes Key:
Deterministic
Stochastic
Constant
Stochastic Dependence
Logical Dependence

Modeling of QT-RR Relationship

As suggested by Hnatkova et al[5], the dynamics between QT and RR intervals are best described by hyper-parabolic functions. Several formulae were considered for QT heart rate correction (Model I - Model VI), before taking into account the circadian rhythm effect, Equation 3.

$$QT_{ij} = \left(QTc_i + Cir_i + \theta_{Pi} * Pla_i + \theta_{Gi} * Gen_i\right) \bullet Corr_i \bullet (1+\varepsilon_{ij}) \quad \text{Equation 3}$$

$$Corr_i = RR_{ij}^{\alpha_i} \quad \text{Model (I)}$$

$$Corr_i = e^{(-RR_{ij}^{\alpha_i})} \quad \text{Model (II)}$$

$$Corr_i = ln(RR_{ij}^{\alpha_i}) \quad \text{Model (III)}$$

$$Corr_i = tan(RR_{ij}^{\alpha_i}) \quad \text{Model (IV)}$$

$$Corr_i = tanh\left(RR_{ij}^{\alpha_i}\right) = \frac{e^{\left(RR_{ij}^{\alpha_i}\right)} - e^{-\left(RR_{ij}^{\alpha_i}\right)}}{e^{\left(RR_{ij}^{\alpha_i}\right)} + e^{-\left(RR_{ij}^{\alpha_i}\right)}} \quad \text{Model (V)}$$

$$Corr_i = arcsin\,h(RR_{ij}^{\alpha_i}) = ln\left(RR_{ij}^{\alpha_i} + \sqrt{\left(RR_{ij}^{\alpha_i}\right)^2 + 1}\right) \quad \text{Model (VI)}$$

Modeling of Circadian Rhythm

Number of harmonic regression functions, ranging from a simple cosine function to two harmonics Fourier series, were integrated to the model, Equation 4, to describe the circadian rhythm (diurnal fluctuations) effect.

$$QT_{ij} = \left(QTc_i + Cir_i + \theta_{Pi} * Pla_i + \theta_{Gi} * Gen_i\right) \bullet RR_{ij}^{\alpha_i} \bullet (1+\varepsilon_{ij}) \quad \text{Equation 4}$$

where:

$$Cir_i = A_{1i}\, sin\left(\frac{2\pi}{24} t_{ij}\right) + A_{2i}\, cos\left(\frac{2\pi}{24} t_{ij}\right) + A_{3i}\, sin\left(\frac{2\pi}{\omega_i} t_{ij}\right) + A_{4i}\, cos\left(\frac{2\pi}{\omega_i} t_{ij}\right) \quad \text{Model (VII)}$$

$$Cir_i = A_{1i}\, cos\left(\frac{2\pi}{24}\left(t_{ij} - \varphi_{1i}\right)\right) + A_{2i}\, cos\left(\frac{2\pi}{\omega_i}\left(t_{ij} - \varphi_{2i}\right)\right) \quad \text{Model (VIII)}$$

Model VII is a two harmonics (24 and ω_i hrs) Fourier series expression. A_{1i}, A_{2i}, A_{3i}, A_{4i} and ω_i are individual parameters; tij is the clock time. Model VIII includes two cosine functions with phase shifts (acrophase), ϕ_{1i} and ϕ_{2i}. Although Model VII and VIII could result in mathematically equivalent solutions, they have different parameterizations that provide flexibility to deal with any parameter correlations.

Results

QT-RR Relationship

Model I & Model VI (the power and the inverse hyperbolic sine models) are found to be the best regression models with similar DIC values and performance. However, Model (I) was chosen due to its mathematical simplicity.

Circadian Rhythm

Around 20 sub-models were assessed by setting one or more of model (VII) or model (VIII) parameters, A_{1i}, A_{2i}, A_{3i}, A_{4i}, ϕ_{1i} and/or ϕ_2 to zero and/or ω_i to 12 hrs. Due to the nature of the data used in this analysis, (number of observations, precision, and their spread over the day) the best circadian rhythm model was found to be a two harmonic Fourier series expression, shown in Equation 5.

$$Cir_i = A_{1i}\, sin\left(\frac{2\pi}{24} t\right) + A_{2i}\, cos\left(\frac{2\pi}{24} t\right) + A_{3i}\, sin\left(\frac{2\pi}{12} t\right) \quad \text{Equation 5}$$

Higher harmonic models exhibited high autocorrelation and poor mixing of chains for some of the parameter estimations. The final model is written as follows:

$$QT_{ij} = \begin{pmatrix} QTc_i + \\ A_{1i}\, sin\left(\frac{2\pi}{24} t\right) + A_{2i}\, cos\left(\frac{2\pi}{24} t\right) + A_{3i}\, sin\left(\frac{2\pi}{12} t\right) \\ + \theta_{Pi} * Pla_i + \theta_{Gi} * Gen_i \end{pmatrix} \bullet RR_{ij}^{\alpha_i} \bullet (1+\varepsilon_{ij}) \quad \text{Equation 6}$$

Estimate of model parameters are listed in Table 1. The mean difference between women and men is around 14.7 ms, which is consistence with other publications[5] and initial data exploration. While, the mean placebo effect is around -4.6 ms. while the individual correction exponent, αi, varies from 0.33 to 0.36.

Table 1. Estimate of Model Parameters

Parameters	Estimate (SD)	90% Credible Interval	Inter Individual Variability (IIV)+
Qtc_i (ms)	407.2 (2.55)	(403.0 , 411.4)	14.34
α_i	0.344 (0.009)	(0.330 , 0.358)	0.052
θ_{pi} (ms)	-4.55 (1.20)	(-6.52 , -2.58)	7.51
θ_{Gi} (ms)	14.67 (4.41)	(7.83 , 22.34)	14.42
A_{1i} (ms)	1.52 (1.00)	(-0.13 , 3.16)	5.97
A_{2i} (ms)	-3.06 (0.35)	(-3.65 , -2.48)	NE++
A_{3i} (ms)	-2.43 (0.26)	(-2.86 , -2.00)	NE++
σ (ms)	3.97 (0.05)	(3.88 , 4.06)	-

\+ Expressed as standard deviation (ms)
++ Not estimated

For each parameter, time series history plots showed good mixing of chains (Figure 6). Similarly, kernel density plots exhibited normal marginal posterior distribution (Figure 7). Autocorrelation plots showed minimal correlation within chains except for few chains of random effect parameters. Furthermore, BGR diagnostics demonstrated that interval within-chains (blue line) and pooled interval between chains (green line) converge to stable values and the ratio between them is close to 1 (red line), as demonstrated in Figure 8 for αi.

Figure 6. History Plots for A_1 and α Mean values

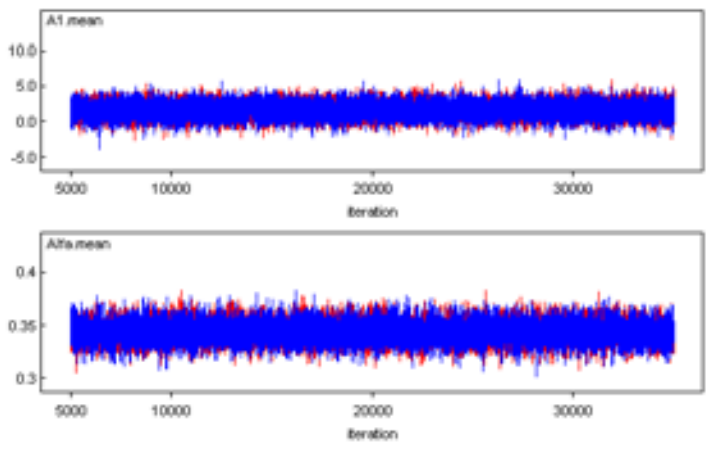
A1.mean
Alfa.mean
iteration

Figure 7. Kernel Density Plots for Fixed Parameters and Deviance

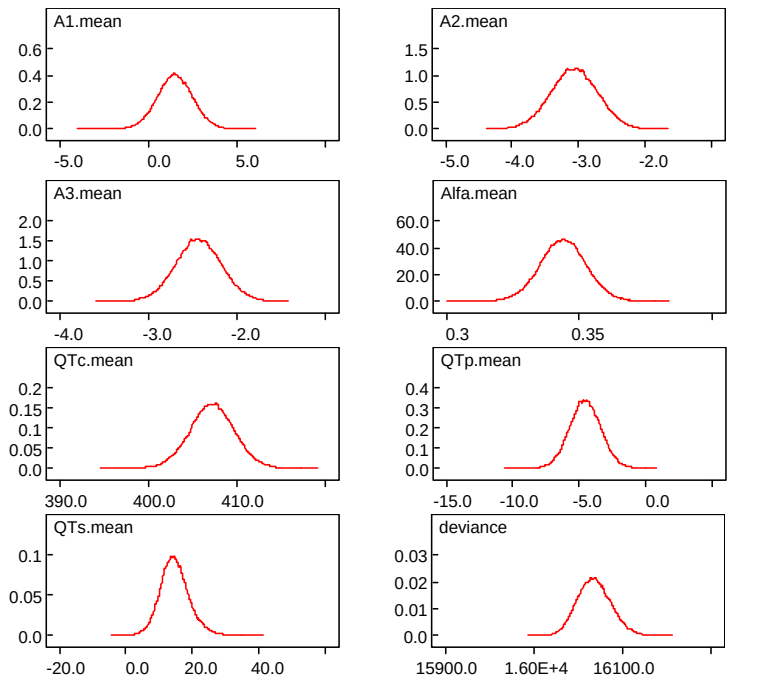
A1.mean
A2.mean
A3.mean
Alfa.mean
QTc.mean
QTp.mean
QTs.mean
deviance

Figure 8. BGR Diagnostics Plot for the Individual Correction Exponent

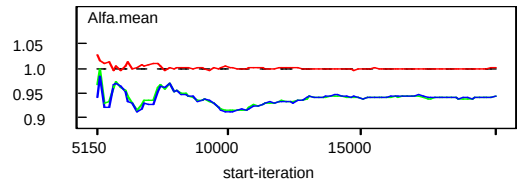
Alfa.mean
start-iteration

Figure 9. Examples of Model Fit for Individual QT Baseline Data

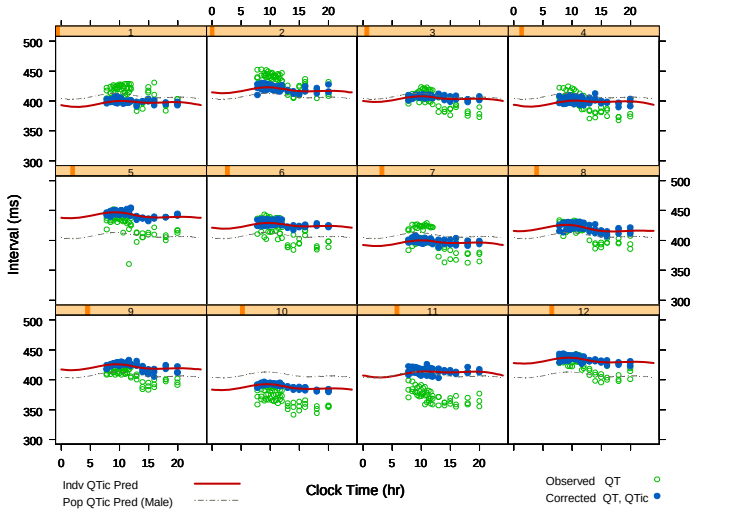
Interval (ms)
Clock Time (hr)
Indv QTic Pred
Pop QTic Pred (Male)
Observed QT
Corrected QT, QTic

Figure 9 shows the time course of the measured QT (open green circles), individually corrected QT, ($QT_{ic} = QT/RR^{\alpha_i}$, filled blue circles), QT_{ic} individual predictions (red solid line), and QT_{ic} population predictions for a typical male (dashed gray line), for 12 subjects. As shown, QT measurements were well described by the final model.

Summary

The present work proposes a framework for modeling baseline QT data using a hierarchical Bayesian approach. The model fit was largely dependent on the method of individualized heart rate correction and circadian rhythm. The performance of periodic functions was tested to appropriately capture diurnal fluctuations in QT and a Fourier series model was selected. Once a model for baseline and placebo data had been established, a variety of concentration-effect models could be tested to characterize the drug effect. The analysis presented here is important in quantifying drug-induced changes in QT interval in drug development.

Acknowledgment

The authors would like to acknowledge Ms. Bhavna Solanki for her help in providing the data.

References

1. Lunn, D.J., Thomas, A., Best, N., and Spiegelhalter, D., WinBUGS - a Bayesian modelling framework: concepts, structure, and extensibility. Statistics and Computing, 10:325—337, 2000

2. Girgis, I.G., Schaible, T., Nandy, P., De Ridder, F., Mathers, J., Mohanty S., Parallel Bayesian Methodology for Population Analysis, PAGE 16 (2007) Abstr 1105 [www.page-meeting.org/?abstract=1105]

3. Piotrovsky, V., Pharmacokinetic-Pharmacodynamic Modeling in the Data Analysis and Interpretation of Drug-induced QT/QTc Prolongation, AAPS Journal. 7(3), E609-E624, 2005

4. Spiegelhalter, D.J., Best, N.G., Carlin, B.P., van der Linde, A., Bayesian measures of model complexity and fit. J. Royal Statist. Society Series B 64(3), 583-639, 2002

5. Hnatkova, K., Malik, M., "Optimum" Formulae for Heart Rate Correction of the QT Interval, Pacing and Clinical Electrophysiology 22 (11) , 1683–1687, 1999

Johnson-Johnson PHARMACEUTICAL RESEARCH & DEVELOPMENT, L.L.C.